Results from GALILEO-1, a first-in-human clinical trial of FLT201 AAV gene therapy in adult patients with Gaucher Disease Type 1

Sharma R[1], Goker-Alpan O[2], Schwartz I[3], Giraldo P[4], Ferrante F[5], Foulds P[5]

1. Salford Royal Hospital, UK; 2. Lysosomal and Rare Disorders Treatment Center, Virginia, USA; 3. Hospital de Clinicas de Porto Alegre, Brazil; 4. Hospital Quironsalud de Zaragoza, Spain; 5. Spur Therapeutics, Stevenage, UK

Poster #318

Introduction and Background

Gaucher Disease

- Gaucher disease (GD) is one of the most common lysosomal storage disorders. Mutations in the *GBA1* gene attenuate or abrogate the activity of the lysosomal acting enzyme glucocerebrosidase (GCase).[1]
- While enzyme replacement therapy (ERT) and substrate reduction therapy (SRT) are currently standard of care for the treatment of type 1 GD (GD1) patients, significant unmet need remains.[2-6]
- Life-long requirements for treatment and variable or incomplete responses negatively impact patient outcomes and quality of life

FLT201

FLT201, an investigational gene therapy for the treatment of GD1, is a novel, proprietary, liver-tropic capsid (AAVS3) with a unique GBA1-85 transgene encoding an engineered variant of β-glucocerebrosidase (GCase85) that provides extended stability in serum and at lysosomal pH compared to wild type GCase, allowing for greater residence time and availability for use by tissues.

Table 1: Stability of GCase85 versus wt GCase (velaglucerase alfa) in *in vitro* analyses

	Human serum Half-life (hours)	Lysosomal pH Half-life (hours)
WT Gase	0.4	6.5
Variant 85	2.4	>144
Fold increase	**6X**	**>21X**

GCase85 has shown:

- similar catalytic properties to human GCase with increased enzymatic stability (**table 1, above**) leading to a continuous presence in both the circulation as well as within cells of interest
- high liver tropism due to the AAVS3 capsid and codon optimization, allowing for lower vector doses, a key component for a favourable safety profile.
- durable expression out to 4 years in non-human primates (study ongoing)
- significant uptake into multiple cells and tissues (liver, spleen, bone, lung, macrophage) and prevention of substrate accumulation across all tested tissues (in Gaucher mice)
- no changes in immunogenicity compared to velaglucerase alfa

GALILEO-1 Study

- A first-in-human, open-label, dose-finding study of a single IV infusion of FLT201. Eligible patients have GD1, are 18 years or older, on stable ERT or SRT for ≥2 years and have a negative AAVS3 neutralizing antibody test.
- Study objectives are to assess safety and tolerability of FLT201 and to investigate the effects on disease-relevant clinical parameters.
- Immune management regimen began 3 weeks post-infusion [prednisone only (n=4), prednisone + tacrolimus (n=2)].
- Discontinuation of background ERT or SRT occurred at the discretion of the investigator once GCase levels were recorded as increased.

Results

Safety (n=6)

Six patients were dosed with FLT201 at 4.5e11vg/kg. **Table 2** (below) details the baseline demographics and disease status for all enrolled patients. All patients demonstrated continued disease activity at study entry.

Table 2: Patient Demographics and Disease Characteristics at Baseline

	Patient 1	Patient 2	Patient 3	Patient 4	Patient 5	Patient 6
Age / Gender	35 / M	25 / M	24 / M	30 / F	24 / M	58 / F
Age at diagnosis	4	3	16	15	20	15
GBA1 Variant	p.Val433Leu; p.Asn409Ser	p.Asn409Ser; p.Leu483Pro	p.Gln112Valfs*32; p.Leu422fs; p.Leu483Pro; p.Ala495Pro	p.Asn409Ser; p.Leu483Pro	p.Asn409Ser; p.Leu29Alafs*18	p.Asn409Ser; p.Trp223Arg
Duration of SoC	4 years	22 years	9 years	14 years	4 years	24 years
Therapy at entry	ERT	SRT	SRT	ERT	SRT	SRT
Plasma GCase activity (μmol/L/h)	0.1	0.09	0.04	<0.1	0.5	<0.1
DBS Lyso-Gb1 (ng/mL)	102.85	10.29	486.41#	72.6	257	52.6
Hemoglobin (g/dL)	15.1	15.2	14.5	13.3	17.0	12.6
Platelet count ($x10^3$/mL)	200	213	124	176	167	113
Spleen volume (MN)	1.88	1.39	2.20	1.73	8.39	5.65
Liver volume (MN)	1.14	1.06	0.81	0.75	1.11	0.88

average of two baseline values

Table 3: Adverse Drug Reactions with 2 or more reports

Summary of ADRs (n≥2)	
Adverse Drug Reactions (ADR)	**# events (# patients)**
FLT201	
Elevated Alanine aminotransferase (ALT)	7 (6)
Fatigue	4 (3)
Activated partial thromboplastin time prolonged	2 (2)
Anti-GCase neutralizing antibodies	2 (2)
Prednisone	
Hyperglycemia	3 (3)
Weight increase	2 (2)
Panic attack	2 (1)
Tacrolimus	
Diarrhea	4 (4)

Figure 1: ALT over time (central lab)

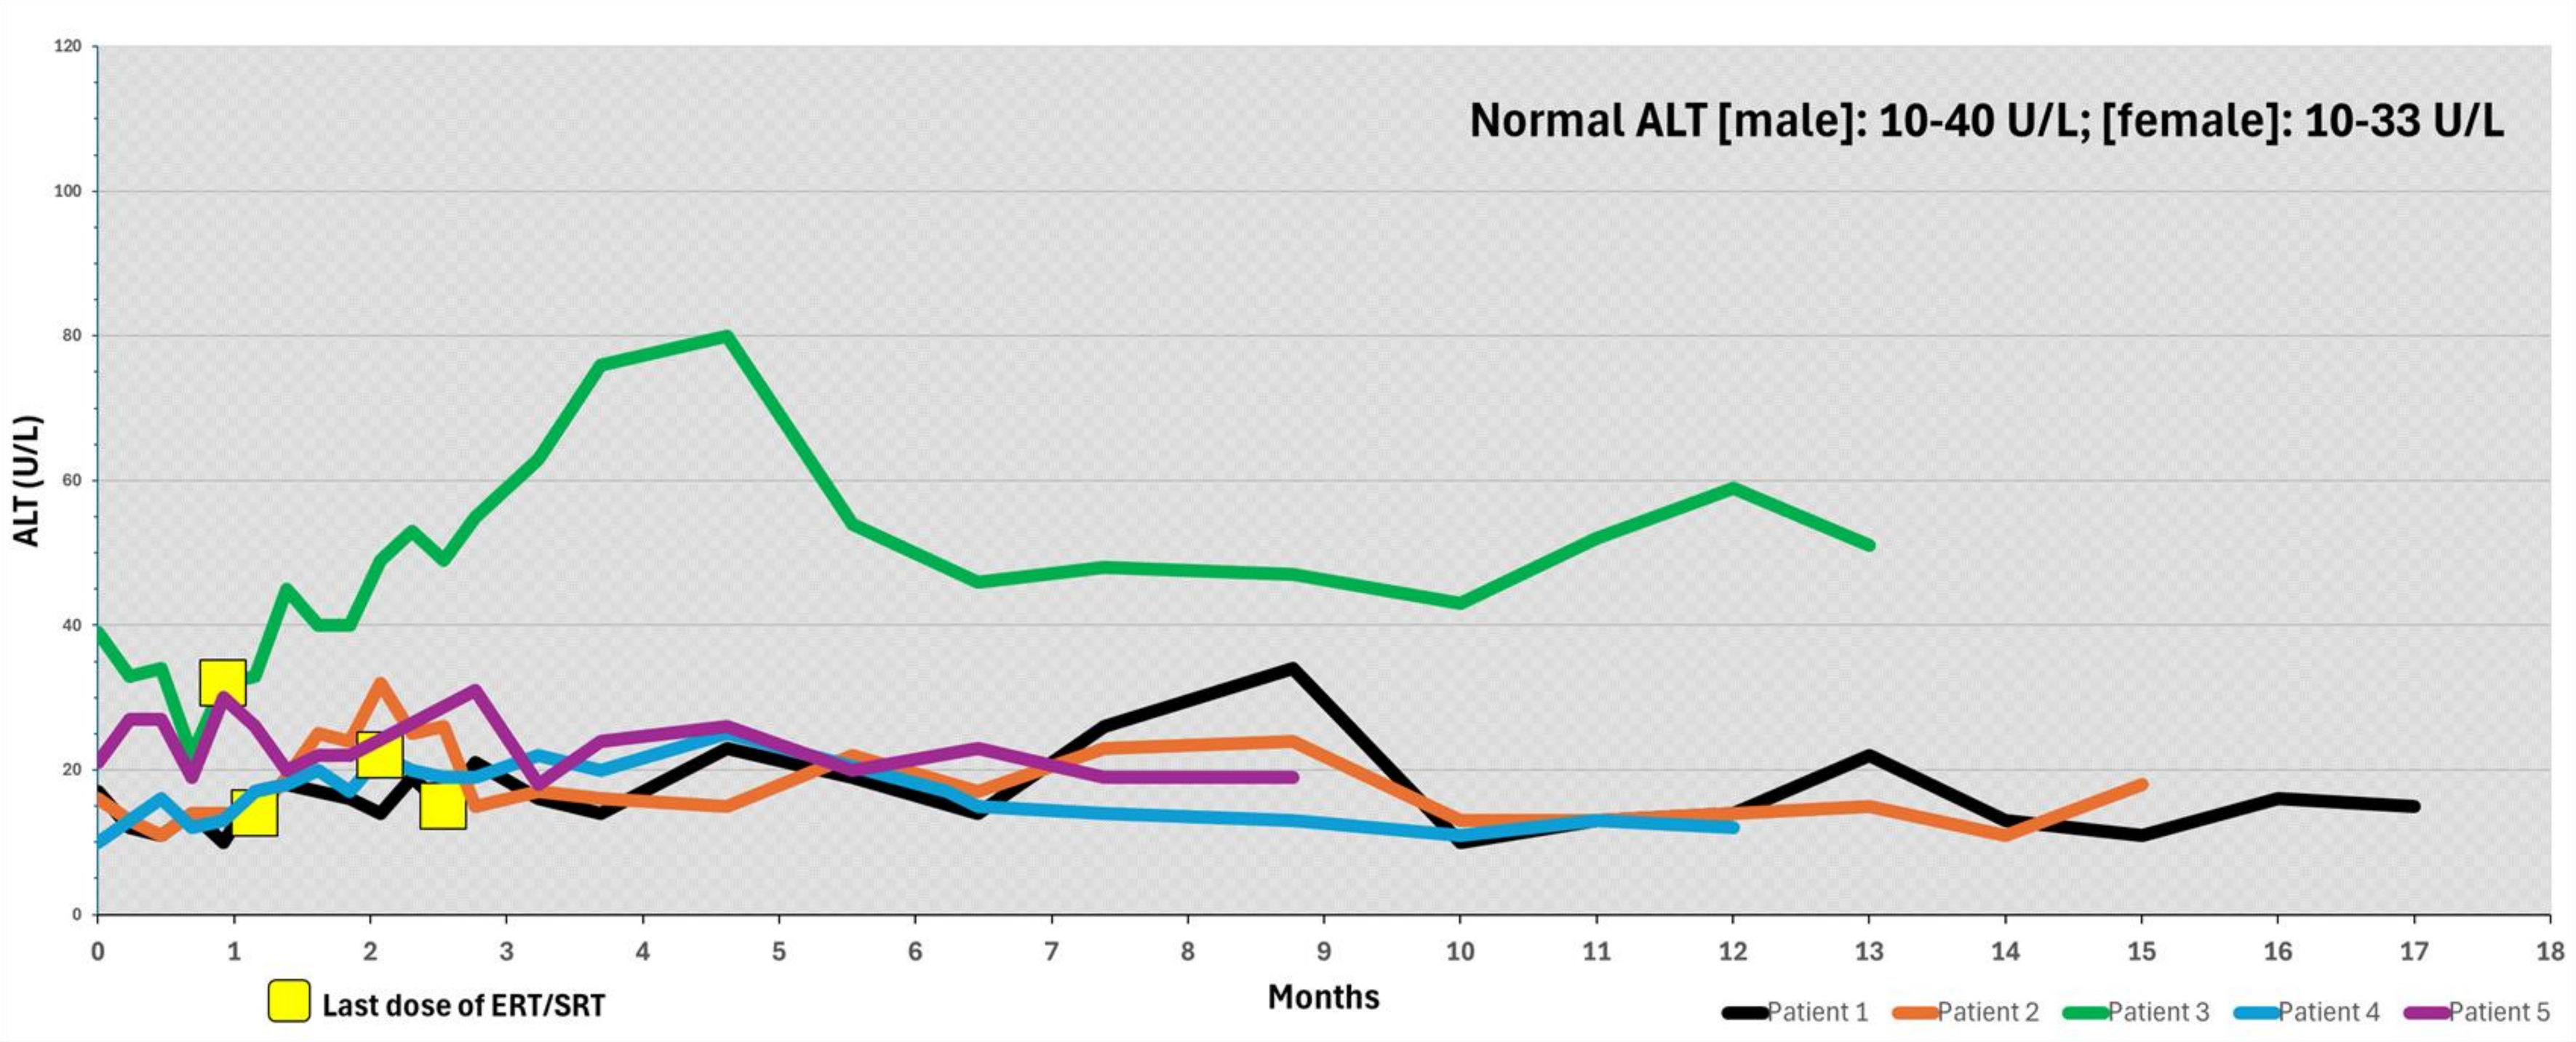
Normal ALT [male]: 10-40 U/L; [female]: 10-33 U/L
ALT (U/L)
Months
Last dose of ERT/SRT
Patient 1
Patient 2
Patient 3
Patient 4
Patient 5

At data cut off, patients have been exposed to the lowest dose of FLT201 (4.5e11vg/kg) for 9-17 months. All patients who came off their prior SoC therapy (between weeks 4-11) remain off (10-14 months). One patient (patient 6), who had a low but detectable anti-AAVS3 titer at baseline, has not had detectable GCase expression and remains on SoC. This patient is not included in the efficacy data set.

Exposure and Safety

All patients experienced at least one ADR. ADRs occurring more than once are listed in **Table 3** (above). The most common ADRs associated with FLT201 were ALT increase and fatigue. ALT elevations even within normal range were considered AEs of special interest as per protocol and, therefore, included as an ADR. Only two patients had an ALT elevation above the normal range and considered related to therapy **(Figure 1, left).** Both resolved, one spontaneously and one after tapering off prednisone.

Two participants developed transient anti-GCase antibodies with no impact on safety. Neither patient experienced loss of clinical benefit during this period although both demonstrated a reduced GCase expression to varying degrees (data not shown), with one (patient 4) rebounding fully to pre-antibody levels once antibody negative. A resulting rebound in GCase measurement in the other patient (patient 5) has not yet occurred.

Efficacy (n=5)

All patients experienced maintenance or improvement in outcomes after receiving FLT201. All but one patient came off their longtime SoC (ERT/SRT) within 4-11 weeks after receiving FLT201 (patient 5 remains on SRT). DBS lyso-Gb1 levels improved rapidly in those with elevated levels at baseline (Baseline mean [range]: 185.8 ng/mL [10.3-486.4 ng/mL]; Week 38 mean/range: 49.8 ng/mL [12.0-172.0 ng/mL], a 73% reduction). Hemoglobin and platelet levels improved or remained in the normal range (**Figures 2 and 3**). One patient experienced a drop in hemoglobin due to a diagnosed iron deficiency. Once iron supplements were initiated, the hemoglobin levels returned to normal. Liver and spleen volumes improved or remained stable (**Figures 4 and 5**). One patient had an enlarged spleen at study entry. By Week 38, this patient had a reduced spleen volume, now into the target goal.

Figure 2: Hemoglobin over time

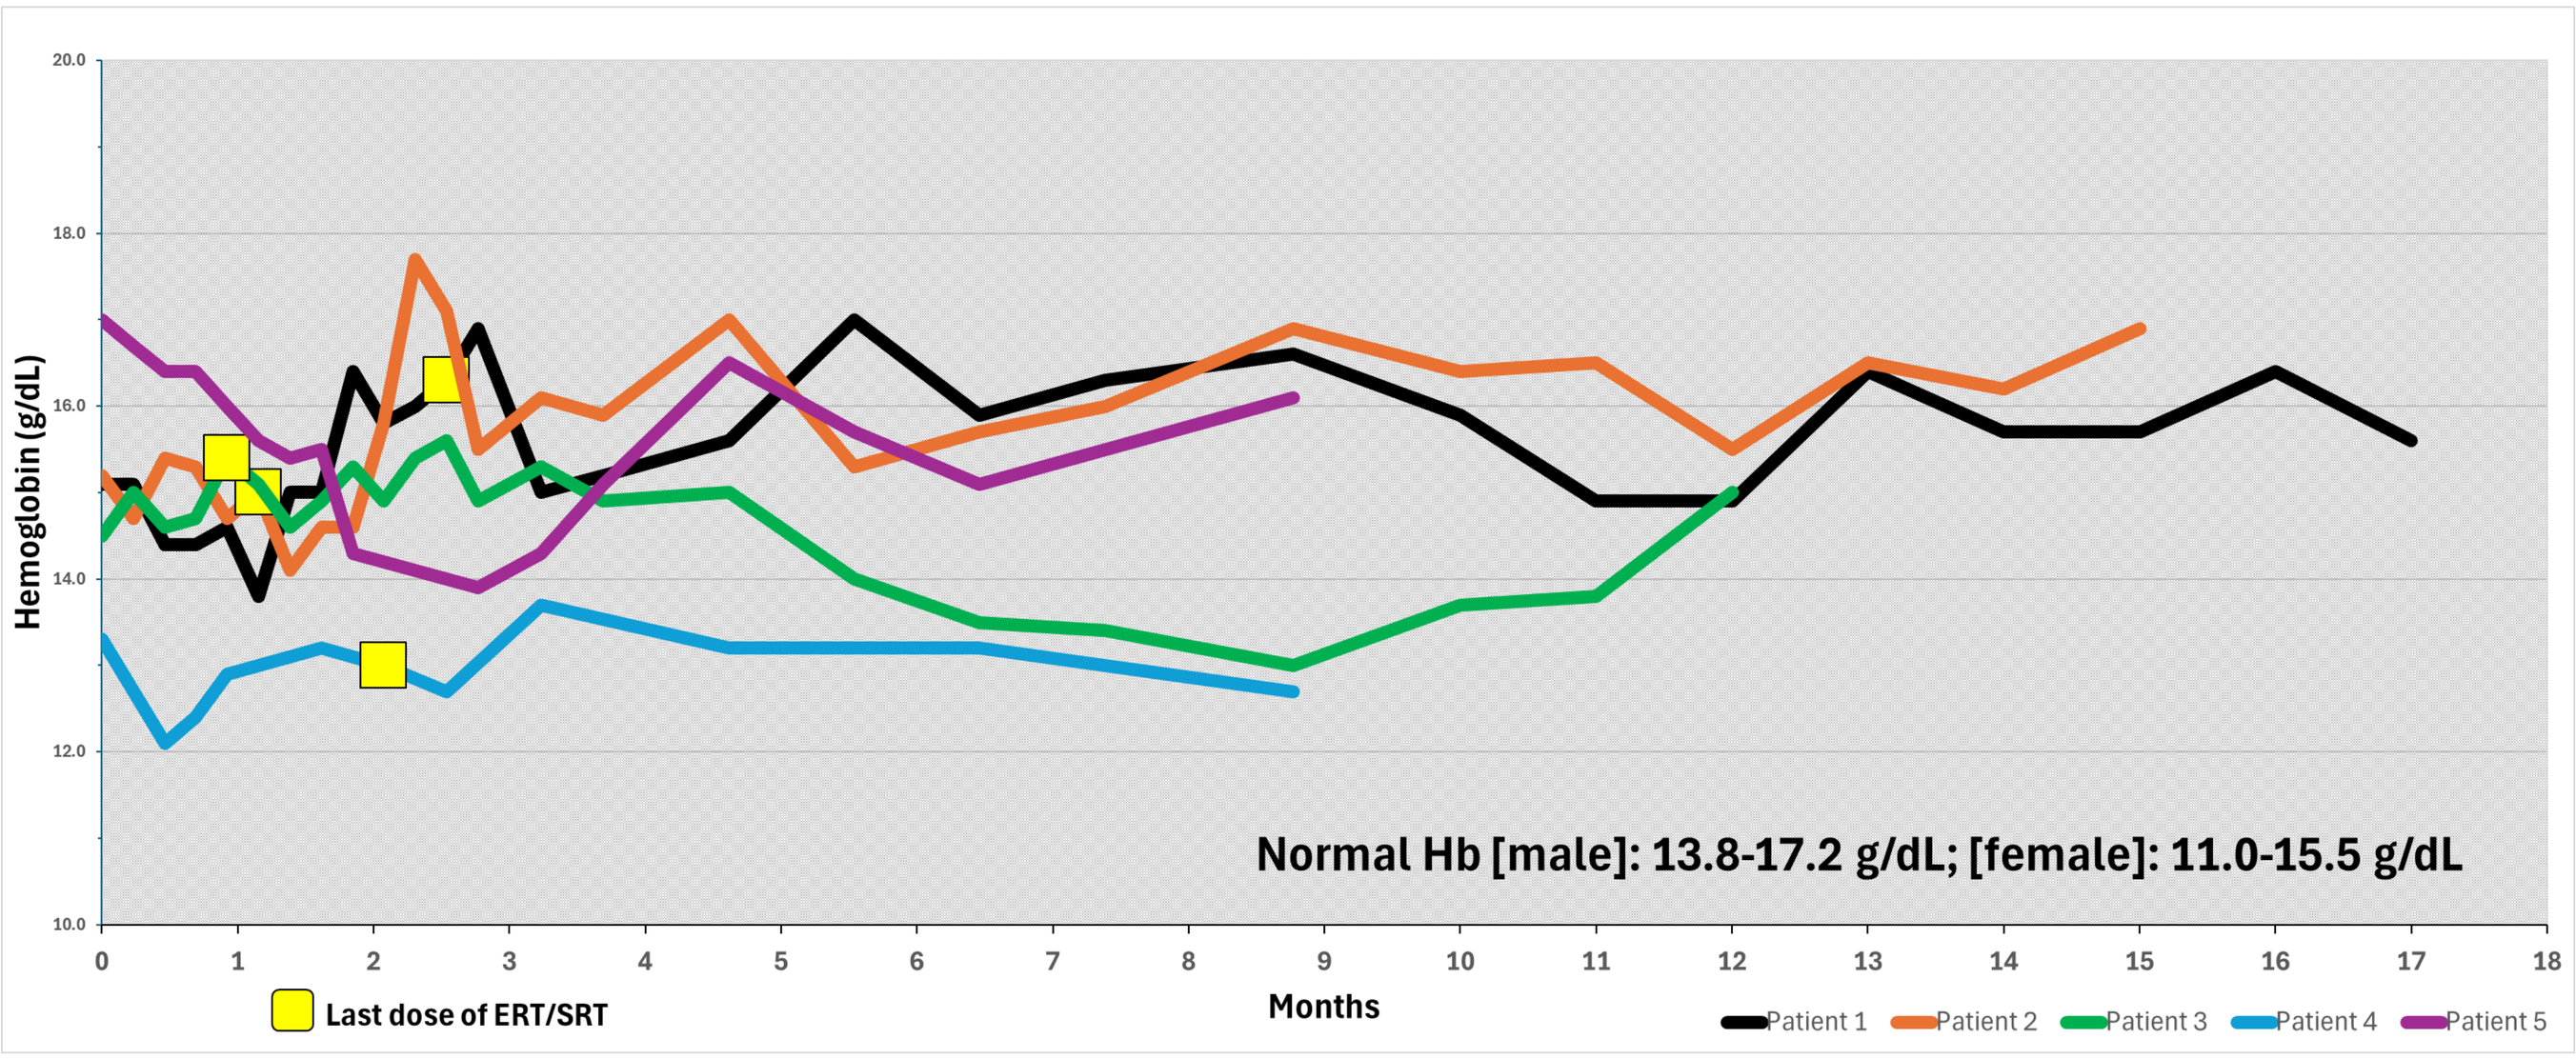
Hemoglobin (g/dL)
Normal Hb [male]: 13.8-17.2 g/dL; [female]: 11.0-15.5 g/dL
Months
Last dose of ERT/SRT
Patient 1
Patient 2
Patient 3
Patient 4
Patient 5

Figure 3: Platelets over time

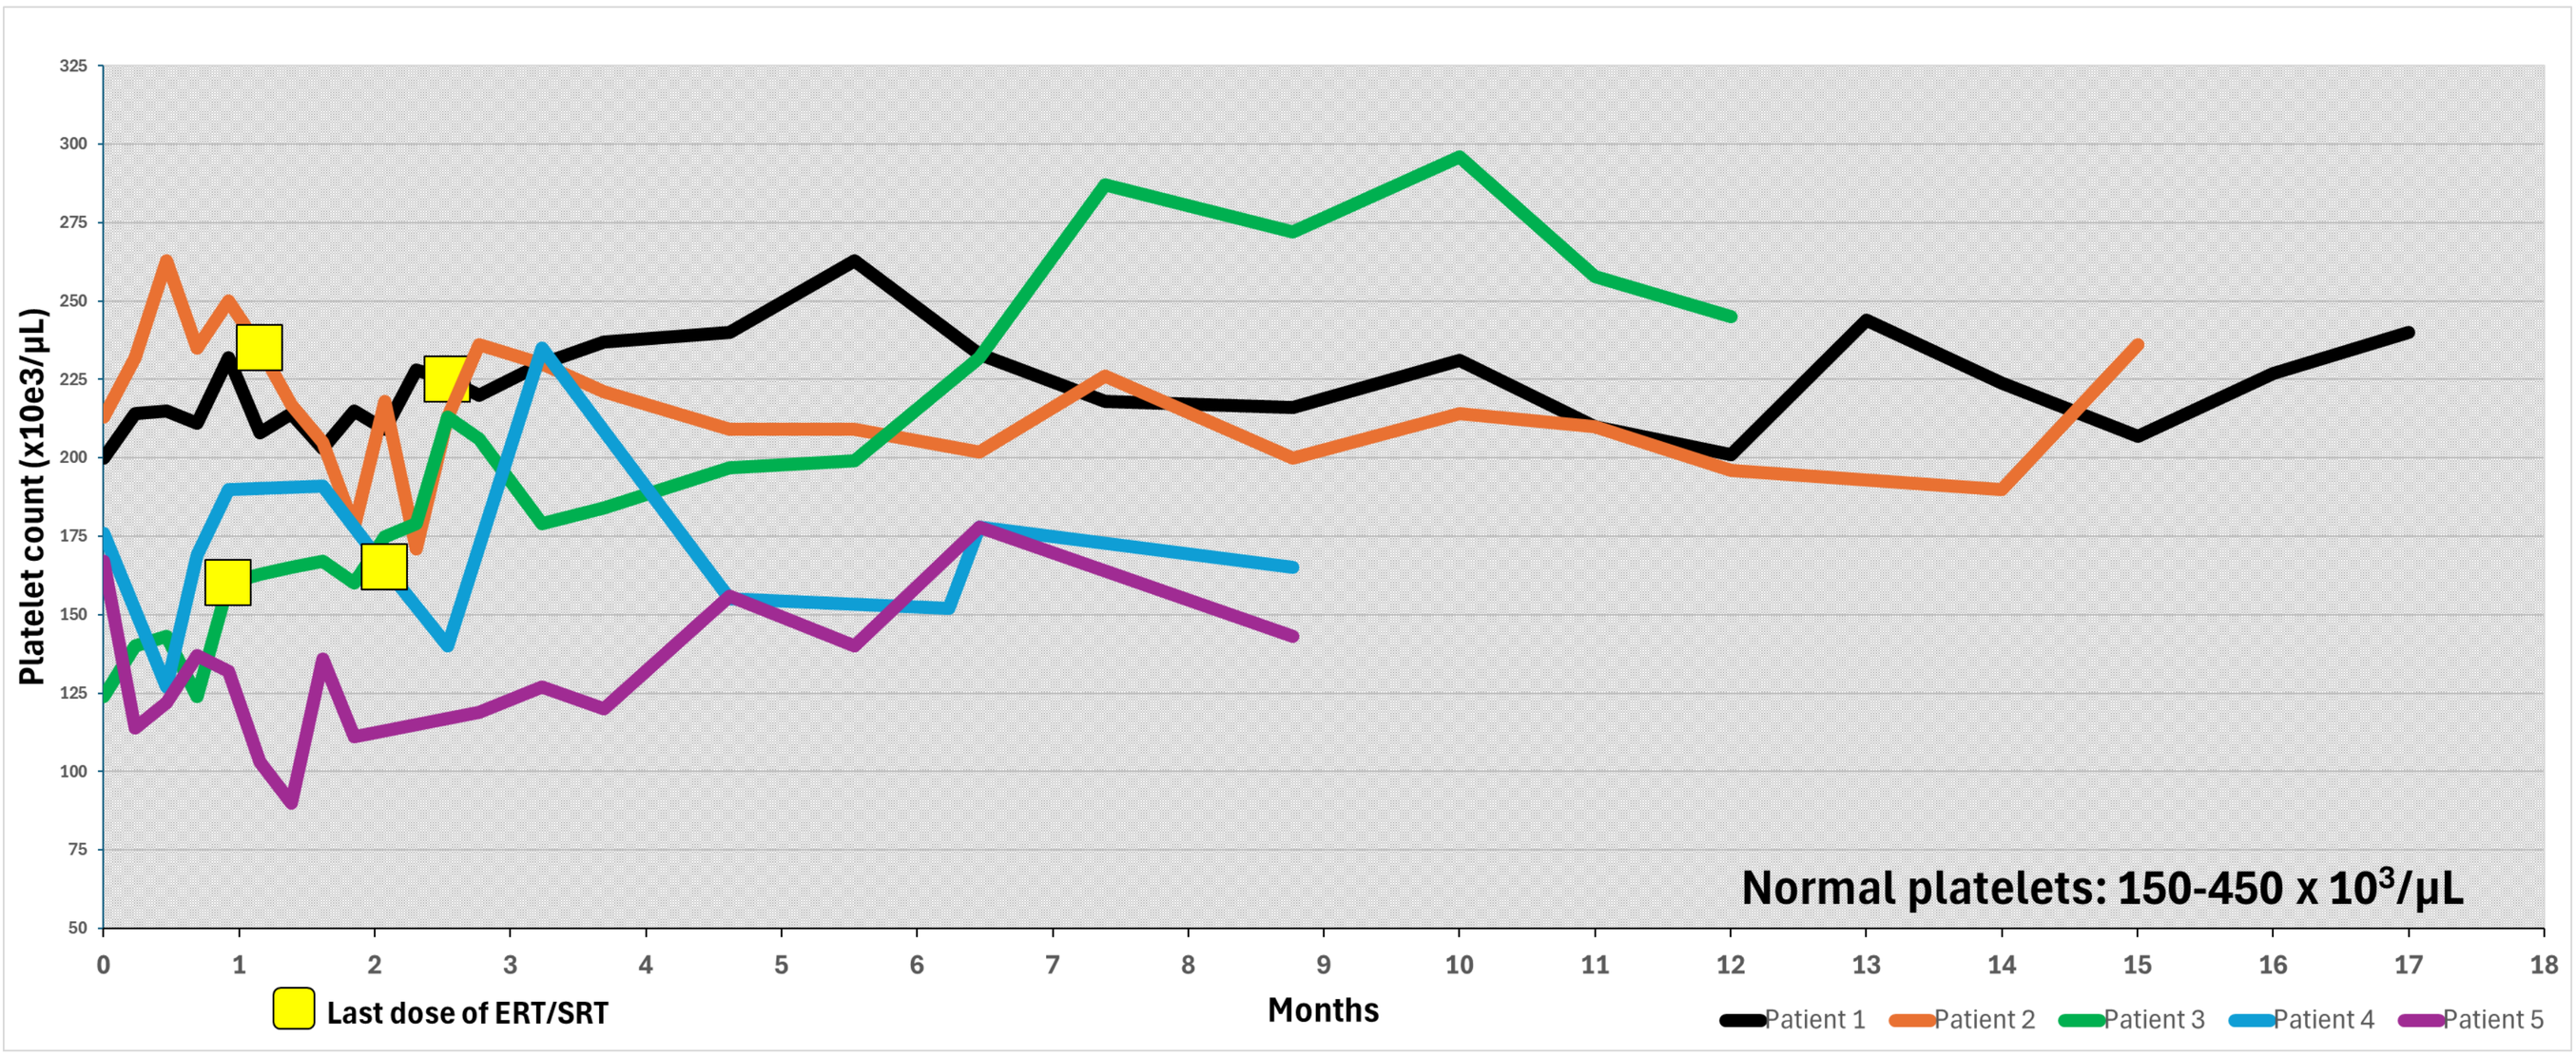
Platelet count (x10e3/μL)
Normal platelets: 150-450 x 10³/μL
Months
Last dose of ERT/SRT
Patient 1
Patient 2
Patient 3
Patient 4
Patient 5

Figure 4:Spleen volume over time (multiples of normal)

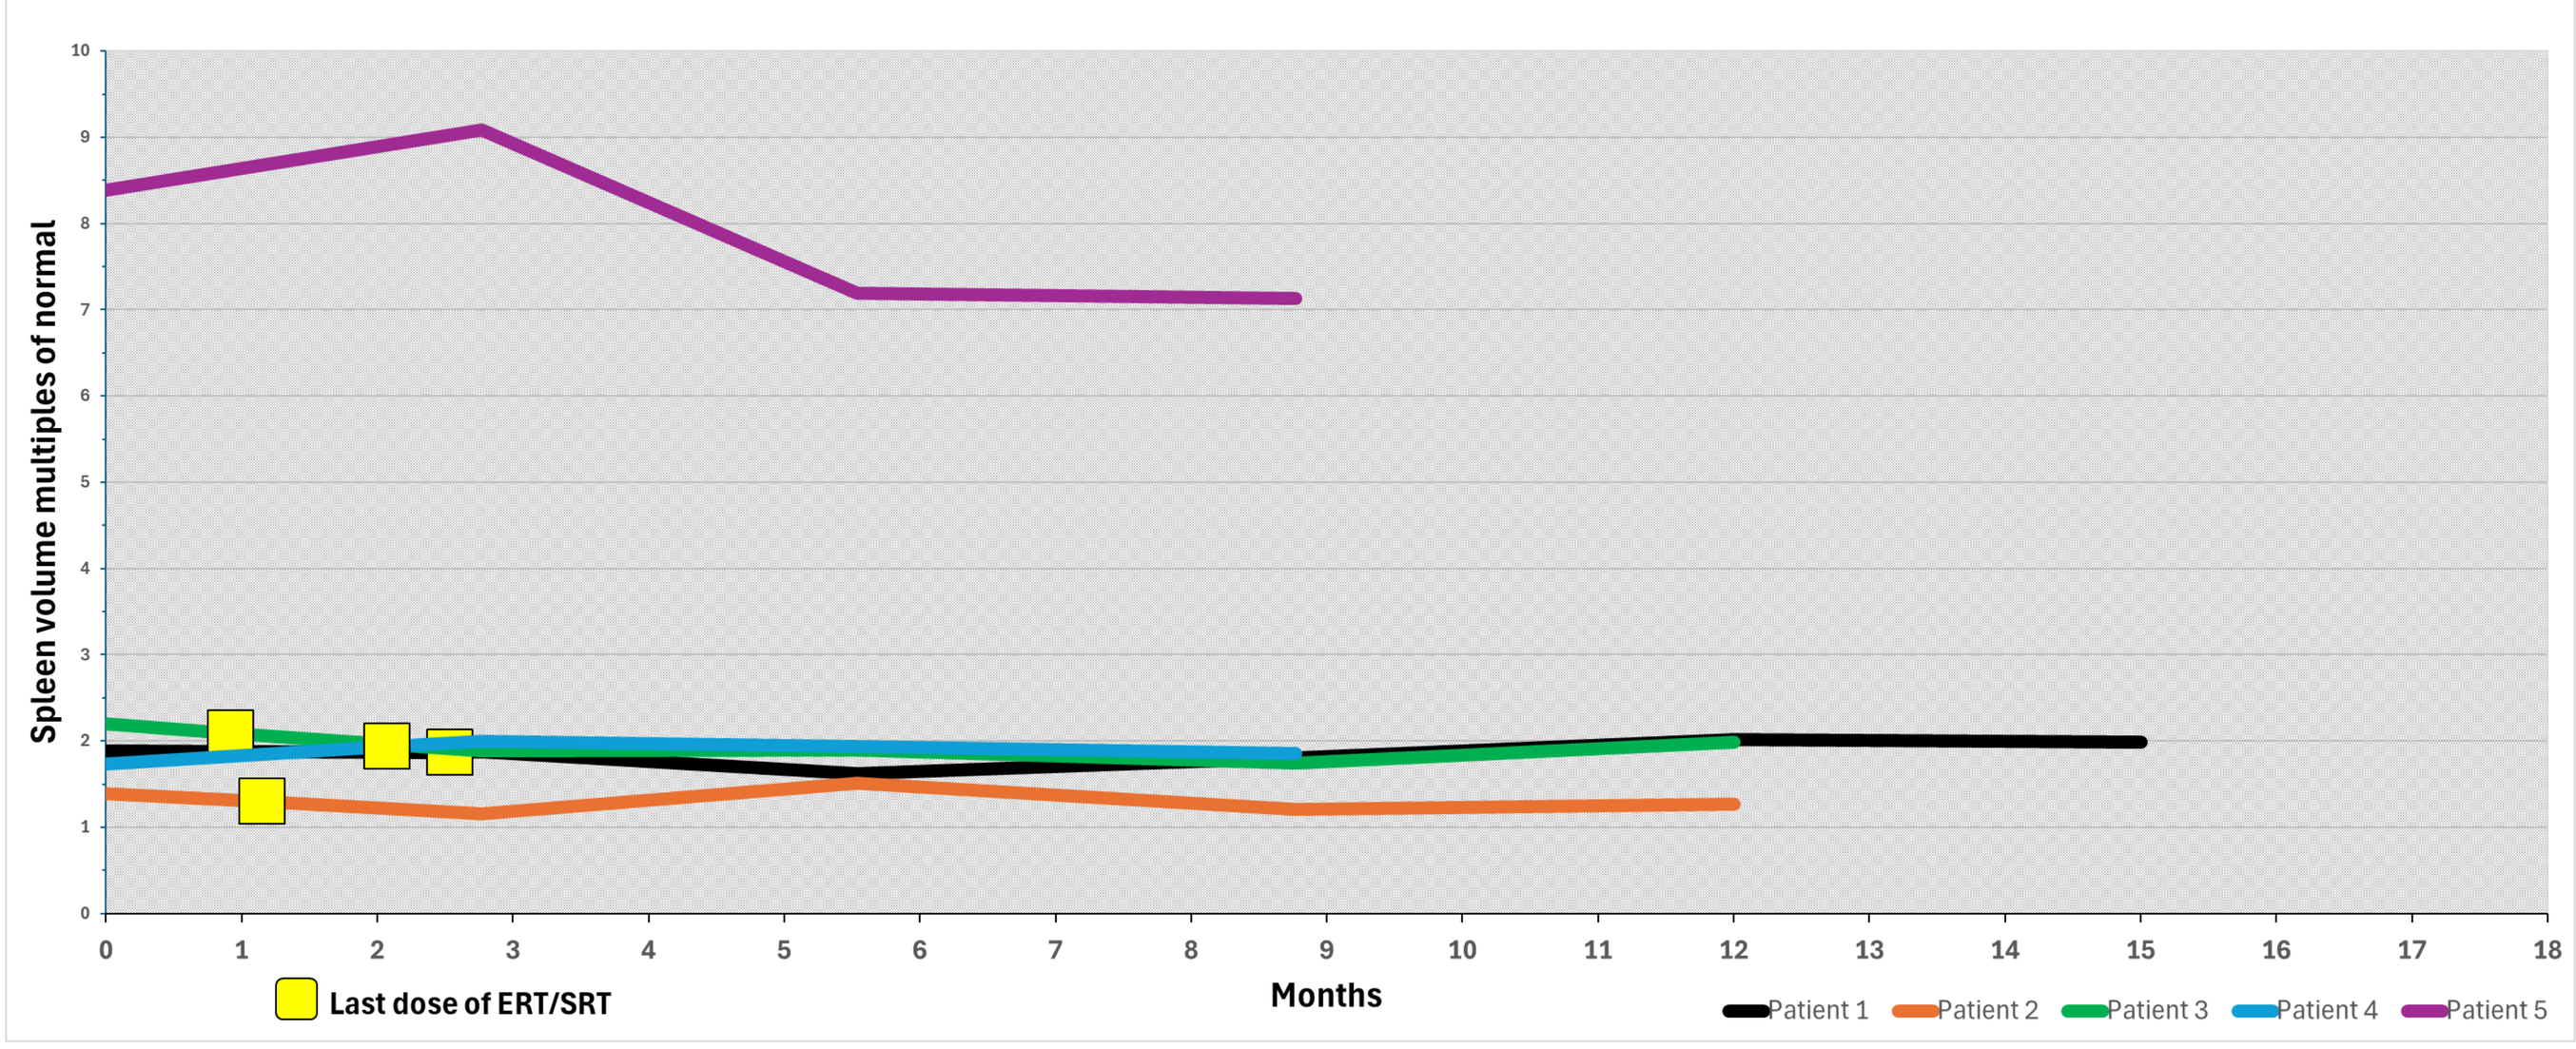
Spleen volume multiples of normal
Months
Last dose of ERT/SRT
Patient 1
Patient 2
Patient 3
Patient 4
Patient 5

Figure 5: Liver volume over time (multiples of normal)

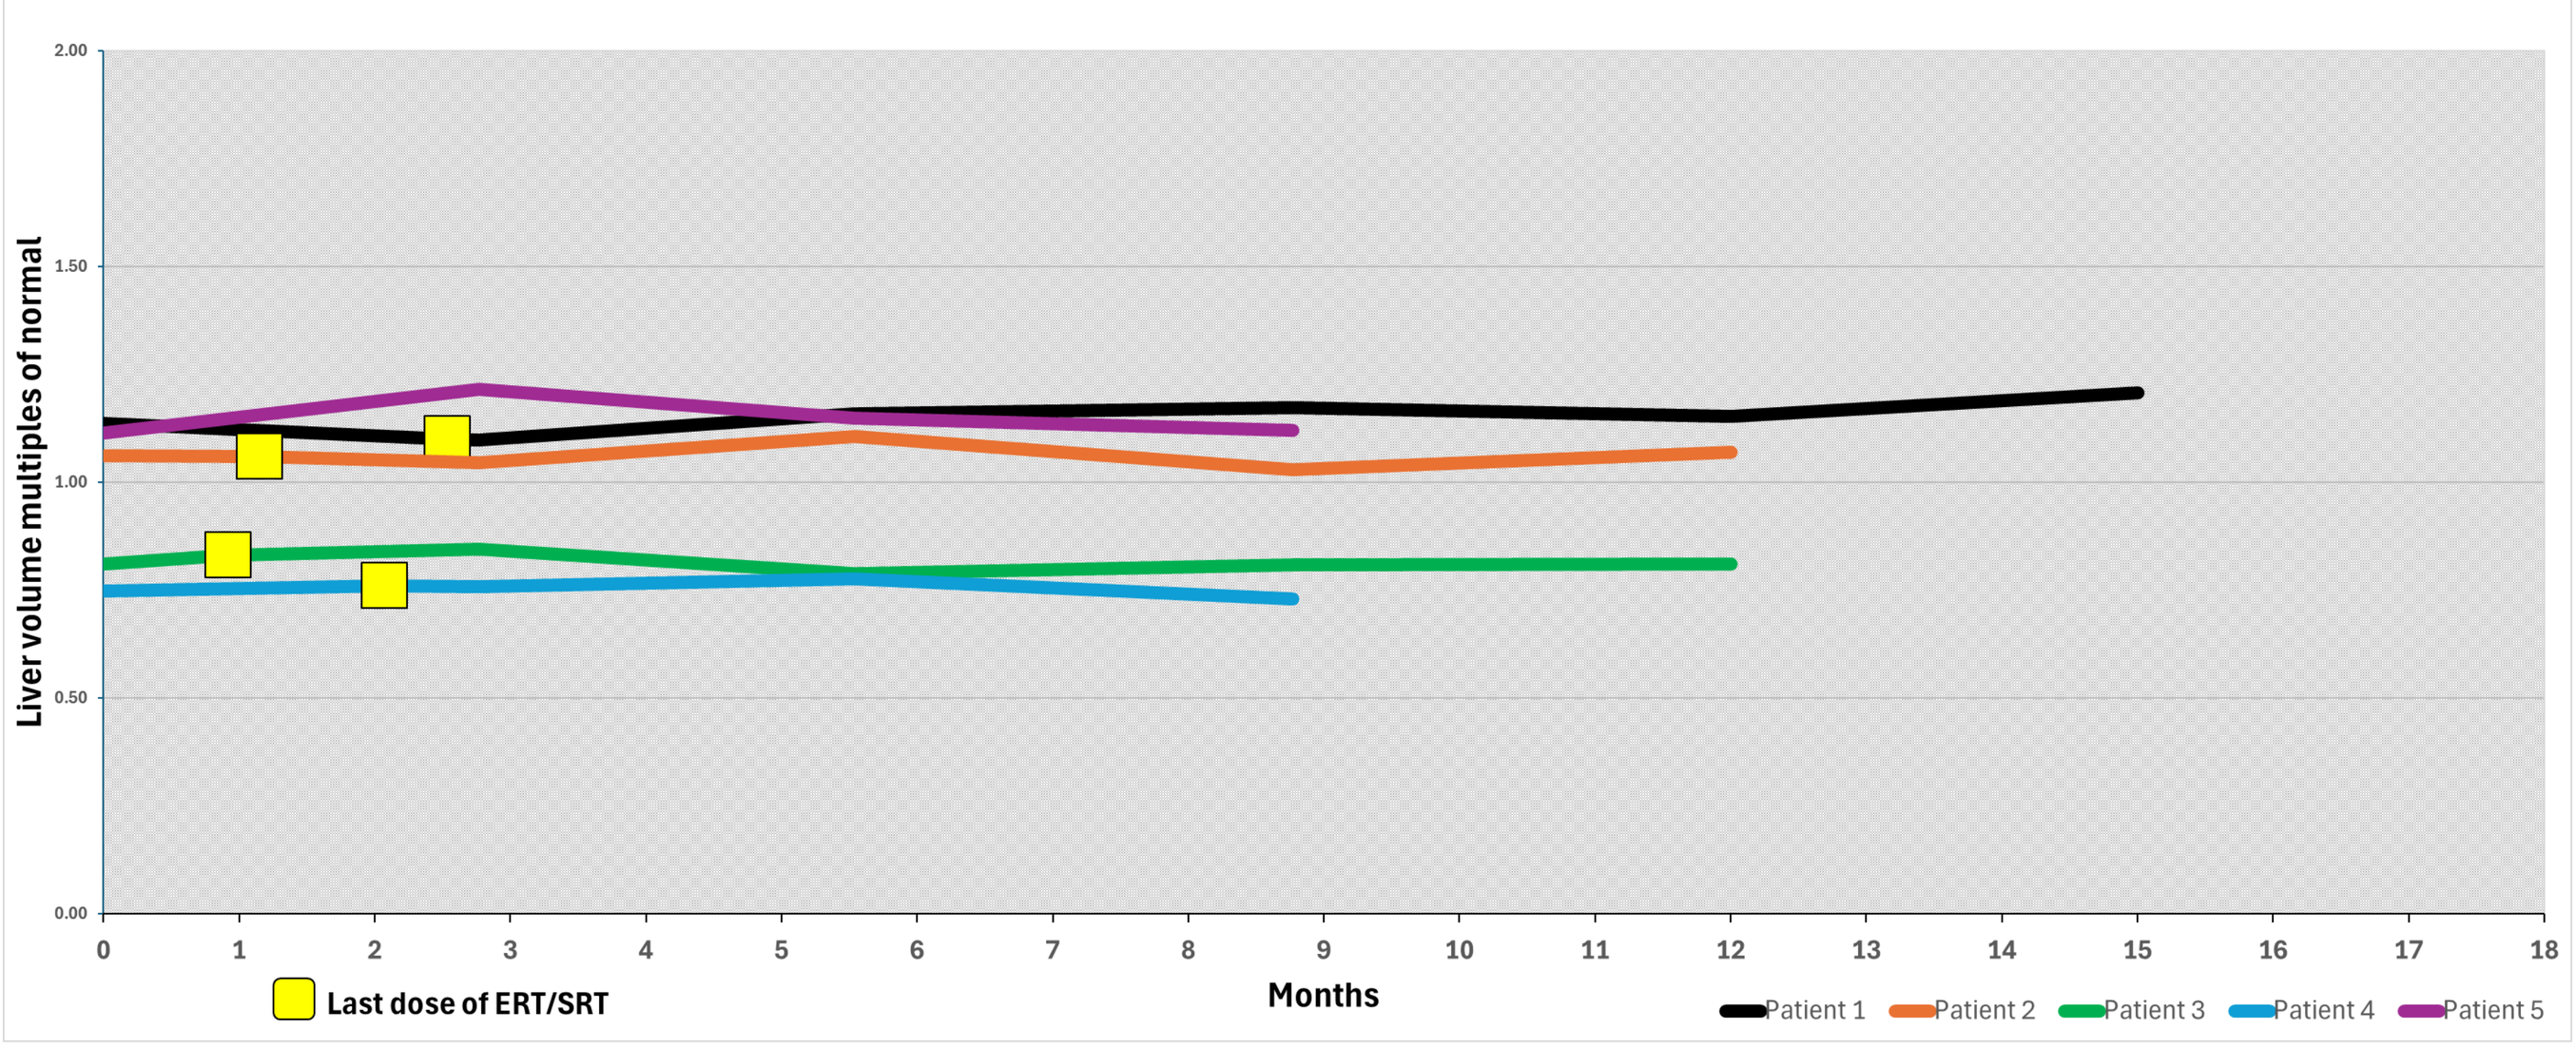
Liver volume multiples of normal
Months
Last dose of ERT/SRT
Patient 1
Patient 2
Patient 3
Patient 4
Patient 5

Conclusions

- **GALILEO-1 trial shows a favorable safety and tolerability profile with a single low dose of 4.5e11 vg/kg.**
- **Continuous expression of GCase85, which is more stable than recombinant human GCase, ensures constant exposure to enzyme.**
- **Clinical parameters and key biomarkers showed sustained improvement or maintenance up to 14 months after withdrawal of ERT/SRT.**
- **FLT201 shows potential for meaningful improvements in clinical outcomes over existing standard of care with a single infusion.**

References

1. Stirnemann J, Belmatoug N, Camou F, et al. *Int J Mol Sci.* 2017;18(2):441. **2.** Gary SE, Ryan E, Steward AM, Sidransky E.*Expert Rev Endocrinol Metab*. 2018;13:107–118. **3.** ShaymanJA. Advances in Gaucher disease: basic and clinical perspectives. Future Medicine Ltd, Grabowski: London; 2013; 240–256.**4**. Weinreb NJ, et al. *J Inherit Metab Dis*. 2013;36: 543–553. **5.**Wyatt K, et al. *Health Technol Assess.* 2012;16:1-543. **6.** Revel-VilkS, et al. *Br J Haematol*. 2018;182:467-480.

Acknowledgements

The authors thank the patients who participated in the GALILEO-1 study and their families, as well as those who support the Gaucher community.

Disclosures: Drs. Foulds and Ferrante are employees of Spur Therapeutics.

Data cut off as of December 06, 2024

Abbreviations: IM: Immune management; ADR: Adverse drug reaction; DBS: dried blood spot; lyso-Gb1: Glucosylsphingosine; MN: Multiples of normal